Epidemiology of Seneca Valley Virus: Identification and Characterization of Isolates from Pigs in the United States

Nick J. Knowles[1], Laura M. Hales[2], Brian H. Jones[2], John G. Landgraf[3], James A. House[4], Kristine L. Skele[2], Kevin D. Burroughs[2] and Paul L. Hallenbeck[2]

1) Institute for Animal Health, Pirbright Laboratory, Ash Road, Pirbright, Woking, Surrey, GU24 0NF, UK.; 2) NeoTropiX, Inc., 351 Phoenixville Pike, Malvern, PA 19355, USA.; 3) Diagnostic Virology Laboratory, National Veterinary Services Laboratories, US Department of Agriculture, Ames, IA 50010, USA.; 4) USDA, Animal and Plant Health Inspection Service, National Veterinary Services Laboratories, Foreign Animal Disease Diagnostic Laboratory, Greenport, NY 11944, USA.

ABSTRACT

Between 1988 and 2005, 12 picorna-like viruses were isolated from pigs showing a variety of clinical symptoms in various locations across the United States. Virus neutralization tests using a specific antiserum raised against one of the isolates showed them to be antigenically related. Six of these isolates were subjected to a pan-picornavirus RT-PCR which employed primer sets targeted to the 3' end of the genome. Sequence analysis of the resulting amplicons revealed all the virus isolates to be closely related to each other and to a newly described picornavirus, Seneca Valley virus (SVV). SVV-001 had originally been isolated from cell culture media and was presumed to have been introduced via bovine serum or porcine trypsin during cultivation of the cells. SVV is a new picornavirus species and most closely related to the cardioviruses; however, it has a number of genome features which have led to the proposal that it be classified in a new picornavirus genus. Based upon the known sequence of SVV, two additional genome regions (VP1 and 2C) of the porcine isolates of SVV were amplified by RT-PCR and their sequences determined. Relationships between the porcine viruses and SVV-001 were the same in all three genome regions studied. Regression analysis indicated that SVV may have been recently introduced into pigs in the USA. Attempts to infect pigs with two of the isolates failed to demonstrate any specific disease. Serological surveys revealed the presence of specific SVV antibodies in pigs, cattle and mice, but not in humans.

INTRODUCTION

Seneca Valley virus SVV-001 was isolated at Genetic Therapy Inc. (Gaithersburg, MD) in 2002 from cell culture media. The complete genome sequence was described by Knowles & Hallenbeck (2005) and shown to be related to, but distinct from, the cardioviruses (Fig. 1). Here we report the biological and serological properties of seven isolates of SVV made from pigs in various locations in the United States (Minnesota, North Carolina, Iowa, New Jersey, Illinois, Louisiana and California) over a period of 14 years. Our epidemiological analyses reveal the presence of neutralizing antibodies in pigs and other farm animals but not in humans. This information, coupled with the isolation of members of SVV in pigs, suggests that pigs are a natural host for SVV.

MATERIALS & METHODS

RT-PCR & DNA sequencing

Total RNA was extracted from 460 µl of virus infected cell culture supernatant using RNeasy™ kits (Qiagen) as per the manufacturer's protocol and resuspended in 50 µl of nuclease-free water. This RNA (5 µl) was used as the template in a one-step RT-PCR (Ready-To-Go™ RT-PCR Beads; Amersham Pharmacia Biotech) as per the manufacturer's instructions. Forward and reverse primer amounts were 20 pmol and 40 pmol, respectively. The complete SVV VP1 gene was amplified using the primers SVV-1C556F and SVV-2A22R located in the VP3 and 2A regions, respectively (Table 1). The mixes were subjected to the following thermal profile: 30 min at 42 °C, 5 min at 94 °C, and then 35 cycles of 1 min at 94 °C, 1 min at 55 °C and 1.5 min at 72 °C, followed by 5 min at 72 °C and held at 4 °C. PCR amplicons were analysed on a 1.5% agarose-TBE gel containing 0.5 µg/ml ethidium bromide. DNA weight markers (GeneRuler 100 bp DNA Ladder Plus, Ready-To-Use; MBI Fermentas) were run alongside the samples to facilitate product identification and quantification. Post-PCR removal of dNTPs and primers was achieved enzymatically using ExoSAP-IT (GE Healthcare) according to the manufacturer's instructions. PCR amplicons were sequenced with the two PCR primer plus six others shown in Table 1 using the DTS Quick Start Kit (Beckman Coulter) according to the manufacturer's instructions. The sequencing reactions were run on a CEQ8000™ Automated Sequencer (Beckman Coulter) according to the manufacturer's instructions.

A pan-picornavirus RT-PCR/sequencing method was used to determine the 3' end of the genome of each virus isolate (Knowles, 2005).

Phylogenetic analyses

Phylogenetic analyses were conducted using MEGA version 3.1 (Kumar et al., 2004). Neighbor-joining trees were constructed using a difference matrix based on the Kimura 2-parameter model of nucleotide substitution. Confidence levels on branches were estimated by bootstrap re-sampling (1000 pseudo-replicates).

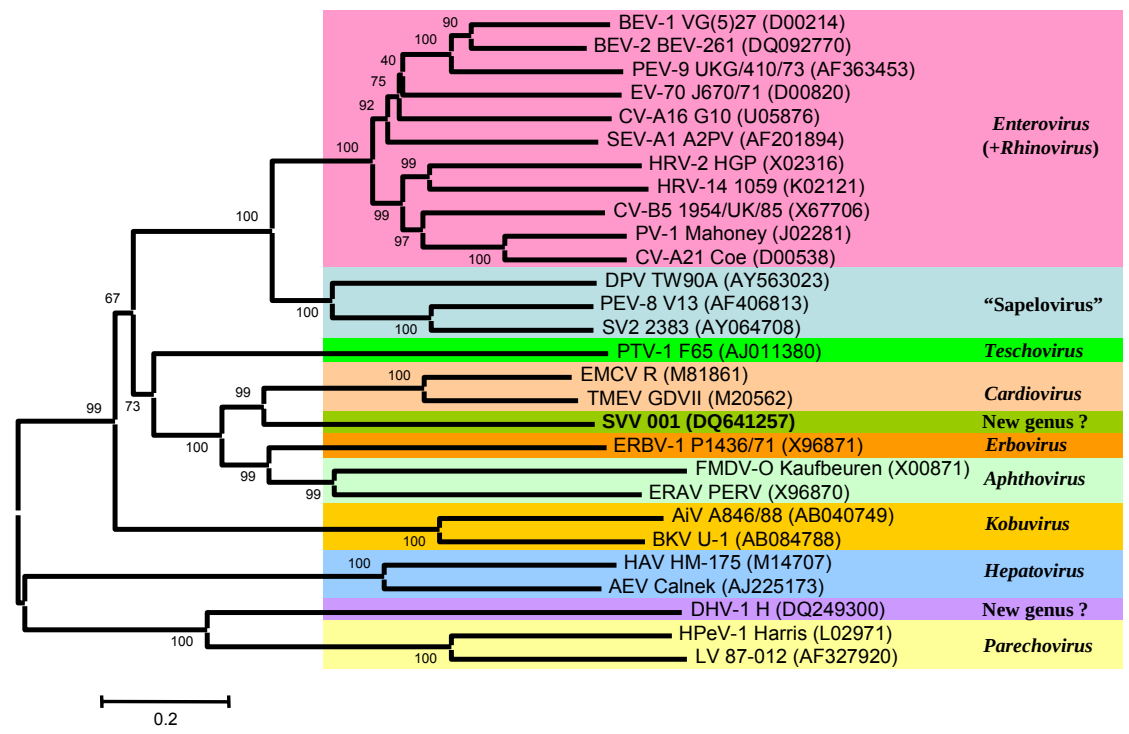
BEV-1 VG(5)27 (D00214)
BEV-2 BEV-261 (DQ092770)
PEV-9 UKG/410/73 (AF363453)
EV-70 J670/71 (D00820)
CV-A16 G10 (U05876)
SEV-A1 A2PV (AF201894)
HRV-2 HGP (X02316)
HRV-14 1059 (K02121)
CV-B5 1954/UK/85 (X67706)
PV-1 Mahoney (J02281)
CV-A21 Coe (D00538)
Enterovirus (+Rhinovirus)
DPV TW90A (AY563023)
PEV-8 V13 (AF406813)
SV2 2383 (AY064708)
"Sapelovirus"
PTV-1 F65 (AJ011380)
Teschovirus
EMCV R (M81861)
TMEV GDVII (M20562)
Cardiovirus
SVV 001 (DQ641257)
New genus ?
ERBV-1 P1436/71 (X96871)
Erbovirus
FMDV-O Kaufbeuren (X00871)
ERAV PERV (X96870)
Aphthovirus
AiV A846/88 (AB040749)
BKV U-1 (AB084788)
Kobuvirus
HAV HM-175 (M14707)
AEV Calnek (AJ225173)
Hepatovirus
DHV-1 H (DQ249300)
New genus ?
HPeV-1 Harris (L02971)
LV 87-012 (AF327920)
Parechovirus
0.2

Fig. 1. P1 aa Neighbor-joining tree, 1000 bootstrap pseudo-replicates

Table 1. Primers used for RT-PCR and sequencing of the VP1 region of the pig Seneca Valley viruses.

Primer name	Primer sequence (5' to 3')	Use
SVV-1C556F	TCGGTTTACTCCGCTGATGGTTGG	PCR, SEQ
SVV-1D187F	GAAGGTGCGCAGCAGGATGAT	SEQ
SVV-1D232R	GCGGGTCGGGAAGCGACTGTTGGGC	SEQ
SVV-1D381F	GGTCTCACTAGAGCCGGATCTGG	SEQ
SVV-1D441R	GGTCGTAGACAAAGCTGGAAGCCTGG	SEQ
SVV-1D563F	CGTCTGTGCTCCCCGTGCGCTGG	SEQ
SVV-1D619R	CCAATCAGCATGCGCCGGTAGACC	SEQ
SVV-2A22R	AGGACCAGGATTGGTCTCGATATC	RT, PCR, SEQ

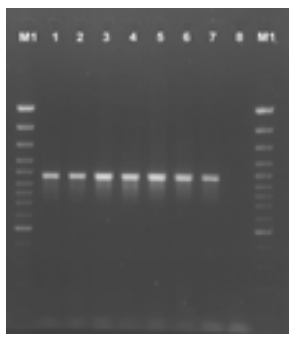
M1 1 2 3 4 5 6 7 8 M1

1. A (NC, 1988)
2. B (MN, 1988)
3. C (IA, 1989)
4. D (NJ, 1990)
5. E (IL, 1992)
6. F (LA, 1997)
7. G (CA, 2001)
8. Negative control (water)

Fig. 2. RT-PCR encompassing the VP1+2A region.

REFERENCES

Knowles, N.J. (2005). A pan-picornavirus RT-PCR: identification of novel picornavirus species. EUROPIC 2005: XIIIth Meeting of the European Study Group on the Molecular Biology of Picornaviruses, Lunteren, The Netherlands, 23-29th May 2005. Abstract A06.

Knowles, N.J. and Hallenbeck, P.L. (2005). A new picornavirus is most closely related to cardioviruses. EUROPIC 2005: XIIIth Meeting of the European Study Group on the Molecular Biology of Picornaviruses, Lunteren, The Netherlands, 23-29th May 2005. Abstract A14.

Kumar, S., Tamura, K. and Nei, M. (2004). MEGA3: Integrated software for Molecular Evolutionary Genetics Analysis and sequence alignment. Brief. Bioinform. 5: 150-163.

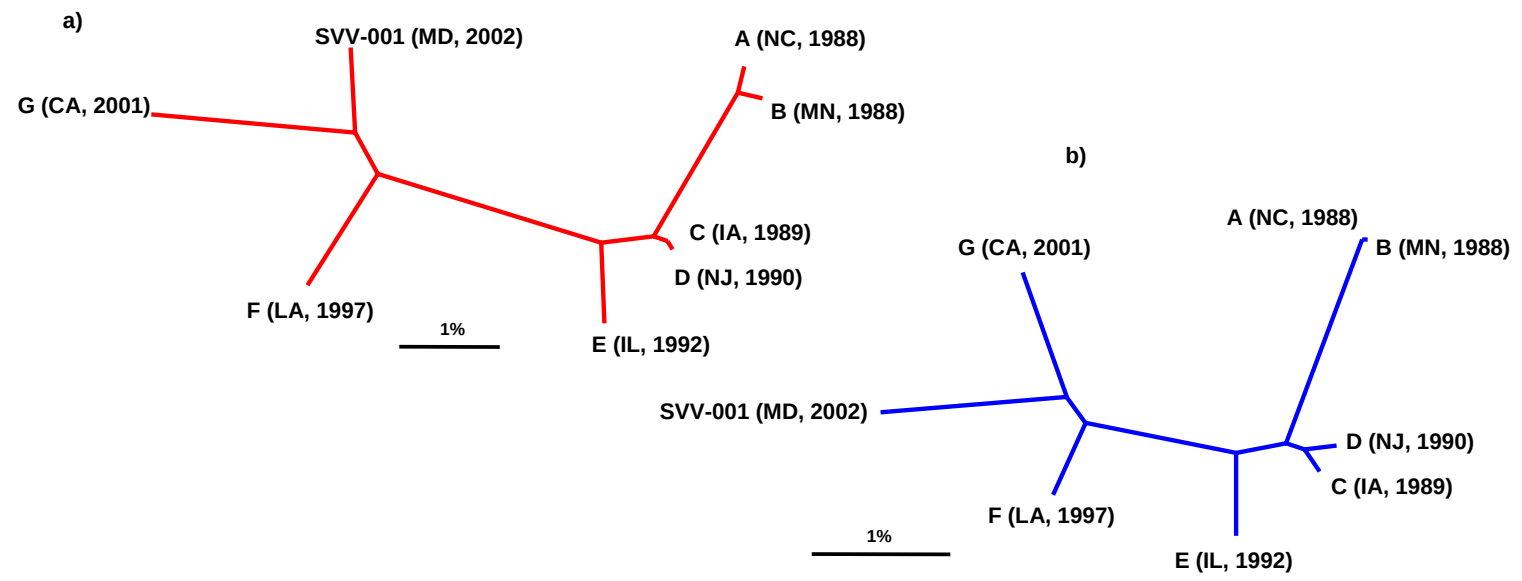
a)
SVV-001 (MD, 2002)
A (NC, 1988)
G (CA, 2001)
B (MN, 1988)
C (IA, 1989)
D (NJ, 1990)
F (LA, 1997)
1%
E (IL, 1992)
b)
A (NC, 1988)
G (CA, 2001)
B (MN, 1988)
SVV-001 (MD, 2002)
D (NJ, 1990)
C (IA, 1989)
F (LA, 1997)
1%
E (IL, 1992)

Fig. 3. Unrooted Neighbor joining trees showing the relationships between SVV-001 and seven of the pig isolates; a) VP1-coding region (798 nt); b) partial 3D[pol] plus complete 3' untranslated region (450 nt).

RELATIONSHIPS BETWEEN SENECA VALLEY VIRUSES

Fig. 3 shows the genetic relationships between SVV-001 and seven of the the SVV isolates made from pigs in the USA. Both the 3' end of the genome and the VP1-coding region gave essentially identical trees. The virus sequences also appeared to fall on an evolutionary time-line. Regression analysis (Fig. 4) confirmed this and suggested that all the viruses had a recent common ancestor, possibly sometime in the early 1980's. Since the viruses were isolated from samples collected in diverse geographic areas of the United States, the results are suggestive of a recent introduction of this virus into the US pig population. Sequence analysis of further isolates may help to confirm this hypothesis. Of particular interest would be isolates from other countries and non-pig species.

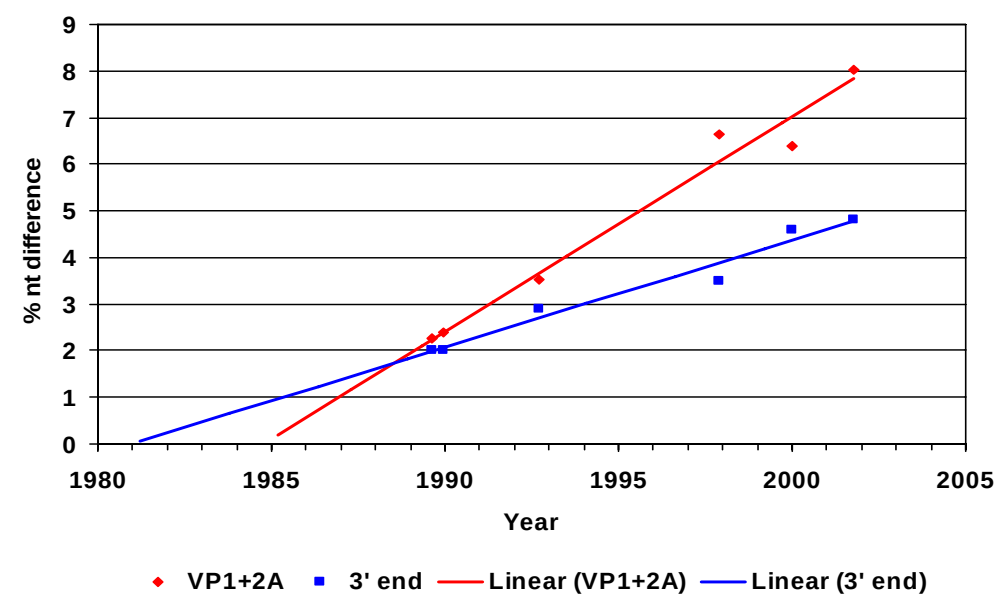
% nt difference
0
1
2
3
4
5
6
7
8
9
1980
1985
1990
1995
2000
2005
Year
VP1+2A
3' end
Linear (VP1+2A)
Linear (3' end)

Fig. 4. Regression analysis of the VP1+partial 2A and 3' end of the genome (using Microsoft Excel). Percentage nucleotide differences to the earliest SVV isolate, A (NC, 1988) are plotted against the date of isolation of each of the later virus isolates.

PREVALENCE OF NEUTRALIZING ANTIBODIES IN FARM ANIMAL POPULATIONS AND OTHER ANIMALS

In order to further study the epidemiology of SVV-001, serum samples were obtained from farm animals including pigs, cows and wild mice. The serum samples were tested individually for the presence of neutralizing antibodies to SVV-001 in a neutralization assay. The presence of such antibodies would suggest that the animals have been exposed and that exposure does not appear to result in overt toxicity. The results are presented in Table 2. The results show that the highest percentage of serum samples that contained neutralizing antibodies came from pigs and cows. This indicates that pigs and other animals are exposed to SVV at levels and in a manner sufficient to induce a measurable immunological response. Additionally, 52 serum samples from four different species of primates were obtained and tested in neutralization assays. None of the samples contained neutralization antibodies to SVV-001.

In previous studies at the USDA, various porcine sera were tested for neutralizing antibodies to SVV isolate F (LA, 1997). Sera were collected in Puerto Rico (4/6 sera had neutralization titres of 1/720 to 1/36,500), New Jersey (2/12 sera had titers of 1/450 and 1/3600), and Texas (2/11 sera had titres of 1/57 to 1/140). This data supports hypothesis that pigs and possibly other farm animals are natural hosts for SVV.

PREVALENCE OF NEUTRALIZING ANTIBODIES IN HUMAN POPULATIONS

In order to assess the exposure of the human population to SVV-001, serum samples were obtained from normal humans, as well as from farmers. More than 60 serum samples from normal humans were screened in a neutralization assay. One sample contained neutralizing antibodies to SVV-001 and had a neutralization titre of 1:8. Serum samples were also obtained from farmers in the Amish population. Fifty samples were tested in a neutralization assay and none contained neutralizing antibodies. This data indicates that human exposure to SVV-001 and SVV-001-like viruses is not prevalent in the human population.

Table 2. Summary of serum samples tested from farm animals in a neutralization assay. Antisera from farm animals were tested in a neutralization assay with SVV-001.

Serum Source	Total no. of animals tested	No. with neutralizing antibodies	%age with neutralizing antibodies	Neutralization titre
Pigs	71	27	38%	1:4
Pigs (from disease-free farm)	30	4	13%	1:4
Cows	30	10	33%	1:8
Mice	35	5	14%	1:2

CONCLUSIONS

We have shown considerable sequence identity between SVV-001 and seven viruses isolated at the USDA from swine farms. Additionally, each of these viruses has been shown to be serologically related to each other, as well as to SVV-001. The selectivity profile of one virus, SVV B (MN, 1988), is identical to that of SVV-001 on all five cell lines tested (data not shown). It is clear that SVV is found in pigs and infection may be common in the United States, however, phylogenetic studies suggest that the virus may have only recently been introduced into this population. It is possible that SVV exists in pigs elsewhere in the world, or perhaps the virus has crossed from another species. It is tempting to speculate a rodent origin, since SVV's closest relatives, the cardioviruses, are viruses of rodents. Because these viruses are genetically, serologically and functionally related, SVV-001, along with these viruses isolated from pigs, will form a new species in the family *Picornaviridae*.